Dopaminergic modulation of executive control function in healthy subjects

Hans Scheffel (1), Stephan Weiland (1), Daniela Roesch-Ely (1), Markus Schwaninger (2), Hans-Peter Hundemer (3), Thomas Kolter (3), Matthias Weisbrod (1)

Departments of Psychiatry (1) and Neurology (2) of the University of Heidelberg, Germany
and Lilly Deutschland GmbH (3)

Background

Executive control is assumed to be a function related to the prefrontal cortex (PFC) [1, 2]. Studies suggest that dopamine influences PFC-functions like working memory, especially via D1-subtype receptors, which predominate in the PFC [3, 5]. Up to now only few studies were conducted to show the participation of dopamine on executive function.
The aim of the study is to investigate the effect of D1-agonists on executive control in healthy subjects. Currently there is no selective D1-agonist available for human research. Therefore a substraction method is used to compare the D1/D2-agonist pergolide with the selective D2-agonist bromocriptine and placebo. Using a double-blind cross-over design, subjects underwent these treatments performing a lateralised Stroop Task.
We predict that: in comparison to bromocriptine and placebo, pergolide increases the executive control function in the left hemisphere (LH), by presenting lower reaction times in the incongruent condition and therefore decreased stroop effect.

Methods

Subjects: 38 healthy young male adults without any somatic or psychiatric diseases. There were no significant differences in age, education and handedness.

Task: In the Stroop Task subjects have to focus their attention selectively on a stimulus in the presence of a more dominant (automatic) stimulus. This ability involves executive function.
The Stroop paradigm was used as a modified computer version of the classic Stroop task. There were 3 different types of experimental conditions: neutral (a row of coloured X), congruent (a colourword and ink are the same) and incongruent (a colourword is printed in a different colour). In each condition the subjects had to name the colour of the words as fast as possible. Reaction time was measured by microphone input. Errors were assessed by the experimenter by means of an answersheet. Stroop effect is measured by reaction time of the incongruent condition - neutral condition.

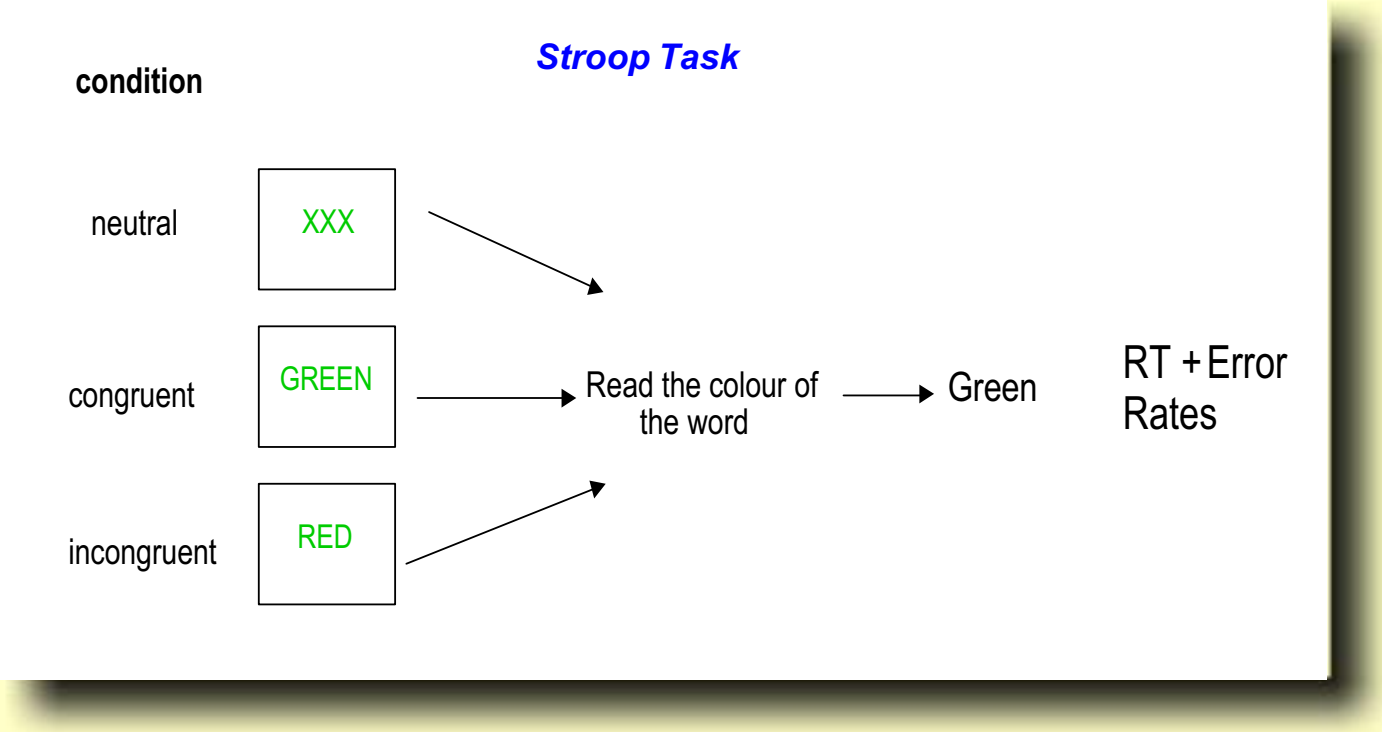
Stroop Task
condition
neutral
XXX
congruent
GREEN
incongruent
RED
Read the colour of the word
Green
RT + Error Rates

Statistical Analysis:
ANOVAs were conducted using reaction times or error rates as dependent variables and medication, experimental condition and laterality as repeated measure variables. Newman-Keuls test was used for post-hoc analysis.

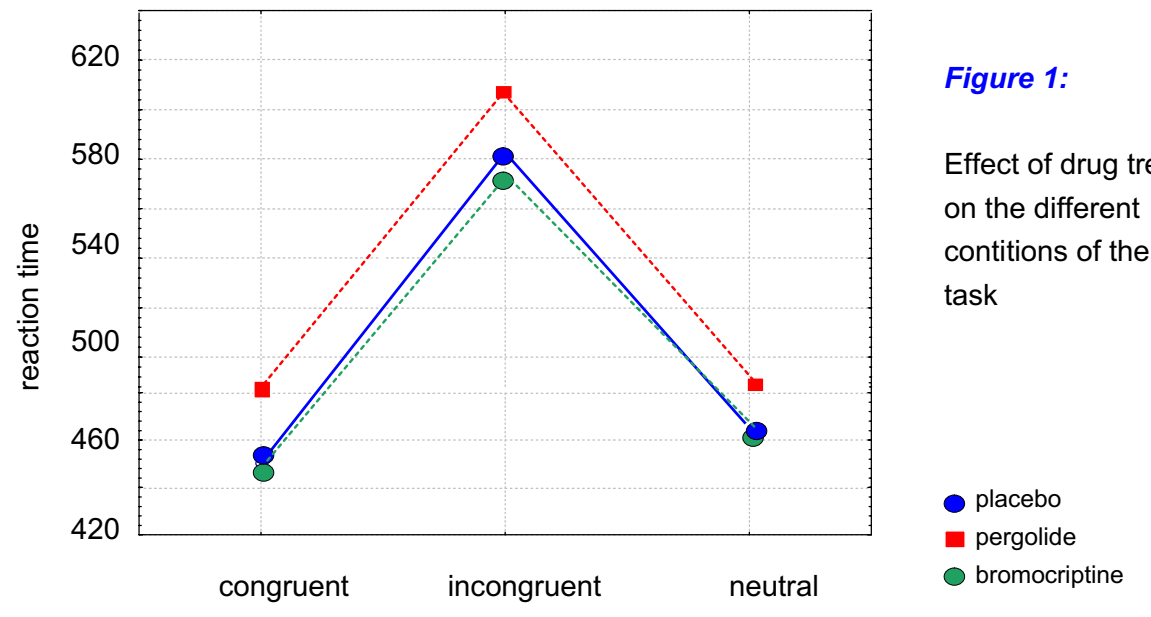
reaction time
620
580
540
500
460
420
congruent
incongruent
neutral
placebo
pergolide
bromocriptine

Figure 1:

Effect of drug treatments on the different contitions of the stroop task

Results

The results showed that:
1) the Stroop Effect is present, that is longer RT in the incongruent condition are found in comparison to the neutral one for all drug types (F(4,148)=2,85;p<0,03) (See Fig.1)
2) subjects showed longer reaction times in all conditions under pergolide medication (See Fig.1)
3) for bromocriptine a decreased stroop effect is found in the left in comparison to the right hemisphere, but it does not reach significant statistical difference (See Fig.2)
4) subjects under bromocriptine showed decreased Stroop effect in comparison to pergolide and placebo in the left hemisphere (F(2,74)=4,38;p<0,02) (See Fig.3)
5) Error rates were very low. More errors were made in the incongruent condition and in the left hemisphere independent of drug type.

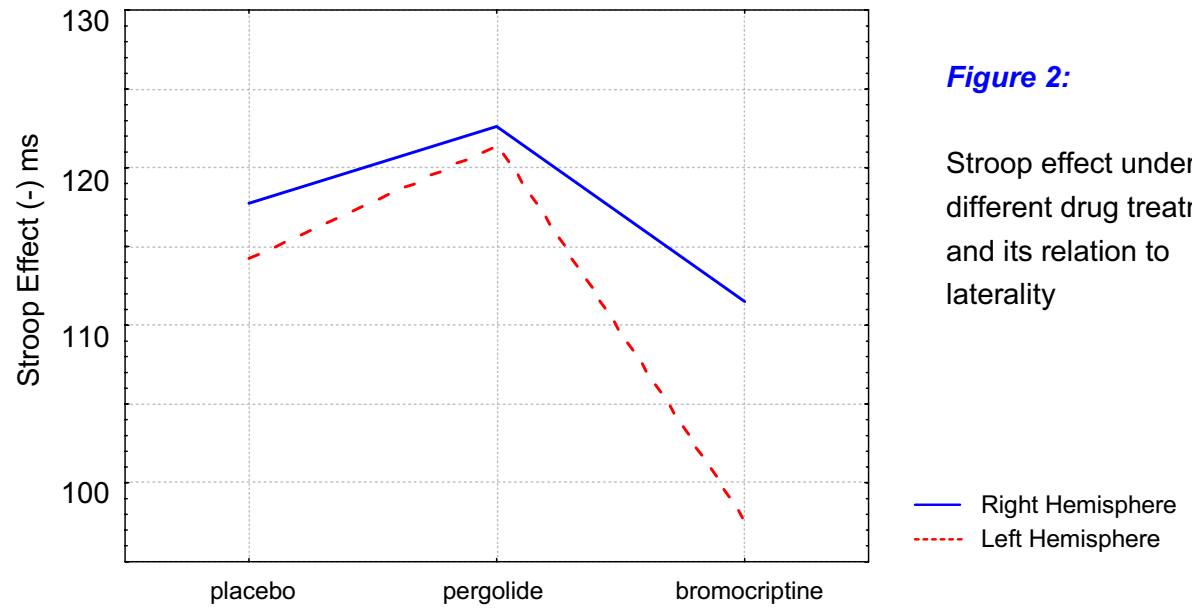
Stroop Effect (-) ms
130
120
110
100
placebo
pergolide
bromocriptine
Right Hemisphere
Left Hemisphere

Figure 2:

Stroop effect under different drug treatments and its relation to laterality

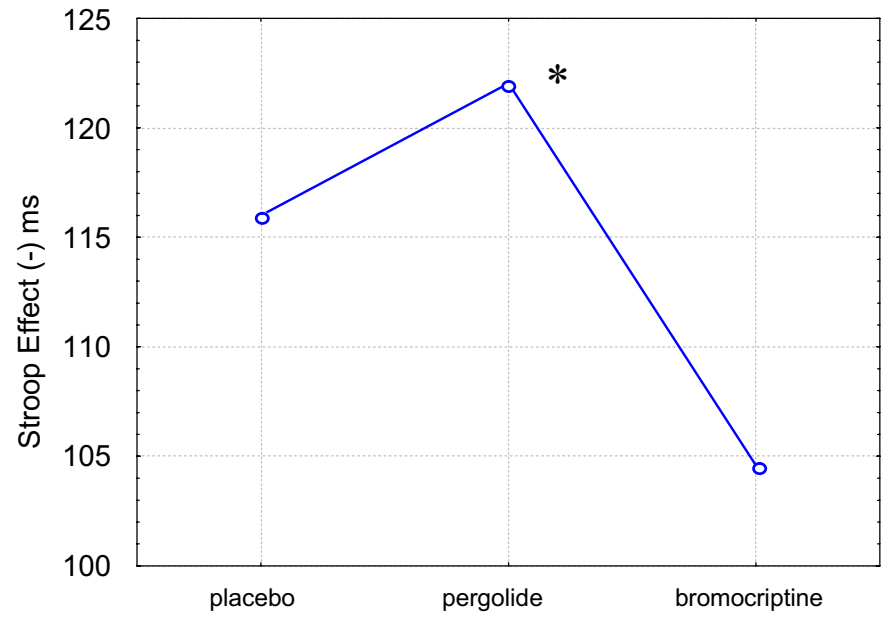
Stroop Effect (-) ms
125
120
115
110
105
100
*
placebo
pergolide
bromocriptine

Figure 3:

Stroop effect under treatments in the left hemisphere

Conclusions

We did not confirm the hypothesis of increased executive function that is, of decreased Stroop effect by subjects under pergolide in comparison to bromocriptine and placebo treatments. Our results point to the modulatory effect of D2-agonists on executive function. There is evidence in the literature [4] which supports the improvement of another PFC function, namely working memory, under bromocriptine in humans. However there is also evidence of a beneficial effect of pergolide in comparison to bromocriptine in a similar task[5]. Reasons for these controversial findings could be due to 1) the different kinds of PFC functions tested, 2) to the not well known receptor interaction of the D1/D2-agonist and its effect on executive and other PFC functions, 3) to the design used (within-subject vs. between-subject design) and its practice effect along sessions. Further studies are needed to clarify these contradictory findings and the differential modulation of dopamine agonists on PFC functions.

Acknowledgements. We thank Dr. Markus Karr for his competent help during subject selection.

References

1. Duncan, J., *Attention, intelligence and frontal lobes*, in *The cognitive neurosciences*, M.S. Gazzaniga, Editor. 1995, MIT Press: Cambridge. p. 721-733.
2. D'Esposito, M., Detre, J.A., Alsop, D.C., Shin, R.K., Atlas, S. & Grossman, M., *The neural basis of the central executive system of working memory.* Nature, 1995. 378: p. 279-281.
3. Sawaguchi, T. and P.S. Goldman-Rakic, *D1 dopamine receptores in prefrontal cortex: involvement in working memory.* Science, 1991. 251: p. 947-950.
4. Luciana, M., *et al.*, *Facilitation of working memory in humans by a D2 dopamine recepetor agonist.* Journal of Cognitive Neuroscience, 1992. 4: p. 58-68.
5. Müller, U., D. Yves and S. Pollmann, D1-Versus D2-receptor modulation of visualspatial working memory in humans. The Journal of Neuroscience, 1998. 18 (7): p. 2720-2728.